Combination of IGF-1R Targeted Alpha Therapy with Olaparib Results in Synergistic Efficacy Against Colorectal and Lung Cancer Xenografts.

M Hu[1], J Forbes[1], R Simms[1], Y Storozhuk[1], J Valliant[1], E Burak[1]
[1]Fusion Pharmaceuticals, Hamilton, Ontario, Canada

Abstract Number: LB130

AACR Annual Meeting 2021

Abstract

Objectives: FPI-1434 is a targeted alpha-particle therapeutic consisting of an IGF-1R targeting antibody radiolabeled with actinium-225. The primary mechanism of action for FPI-1434 is induction of double-stranded DNA breaks (DSB) in targeted tumors resulting in cell death. Poly (ADP-ribose) polymerase (PARP) is part of the cellular mechanism that repairs DSB. In cancer patients with genetic defects in DSB repair (e.g. BRCA1/2), the PARP pathway becomes a primary repair system and its inhibition results in cell death. This mechanism has been leveraged as a therapy against DNA-repair deficient tumors leading to FDA-approval of PARP inhibitors (PARPi) including olaparib. Treatment with PARPi to block repair of DNA damage driven by FPI-1434 may act synergistically to increase the lethal DNA damage load. To that end, Fusion has performed studies to combine FPI-1434 and olaparib against tumor models with no pre-disposing defects in DNA repair.

Methods: Colorectal (Colo-205) or lung (A549) cancer xenografts were established in Balb/c nude mice. For efficacy studies, a dose combination matrix for FPI-1434 and olaparib was tested. FPI-1434 was dosed (i.v.) once at 20-200 nCi followed with olaparib (i.p.) at doses of 0-50 mg/kg. Olaparib was dosed 24h after FPI-1434 using a 5 day on/2 day off schedule (5 mice/group). DSB formation (γH2AX) and apoptosis (cleaved caspase 3) were evaluated in treated tumors by IHC staining.

Results: DSB formation and induction of apoptosis were detected in FPI-1434 treated tumors in a time and dose dependent manner. DSB formation was observed in all areas of tumor containing intact or apoptotic cancer cells confirming FPI-1434 mechanism. In tumor efficacy studies, olaparib had no single-agent efficacy in the Colo-205 or A549 models. Single doses of FPI-1434 at 20 nCi had no effect on Colo-205 tumors, suppressed growth at 50 nCi and caused regression at 100 nCi. Combination efficacy was seen in the 20 and 50 nCi dose groups, including olaparib. Doses of 20 nCi FPI-1434 and 25 mg/kg olaparib had the strongest combined effect in this model. In the A549 model, single doses of FPI-1434 had no effect at 20 or 50 nCi, caused growth suppression at 100 nCi and regression at 200 nCi. Combination efficacy was seen in the 20, 50 and 100 nCi dose groups, including olaparib. Doses of 50 nCi FPI-1434 and 25 mg/kg olaparib had the strongest combined effect in this model. In general, the strongest efficacy was observed by combining ineffective single-agent doses of the two compounds. Strong efficacy of high dose FPI-1434 masked observable combination effects suggesting that FPI-1434 was the therapeutic driver.

Conclusions: Olaparib co-dosing enhanced FPI-1434 efficacy which supports consideration of this combination for clinical use. Ineffective single-agent doses, when combined, resulted in synergistic efficacy in both Colo-205 and A549 models suggesting that the mechanism can be applied to multiple cancer types where predisposing mutations in DNA repair are lacking. The strongest combination effect appeared to occur at the low single-agent doses suggesting that PARP inhibition may potentiate efficacy at lower clinical doses of FPI-1434.

Background

Type I insulin-like growth factor receptor (IGF-1R) is a transmembrane protein which is overexpressed in solid tumors including HNSCC, gynecological cancers, and Her2-negative breast cancers. FPI-1434 is a radioimmunoconjugate consisting of a humanized monoclonal antibody that binds to the external domain of IGF-1R, a proprietary bifunctional chelate, and the alpha-emitting radionuclide actinium-225 (Ac-225). Internalization of the radioimmunoconjugate and decay of Ac-225 causes tumor cell death primarily through induction of double-stranded DNA breaks.

IGF-1R is expressed on nearly all tumor types

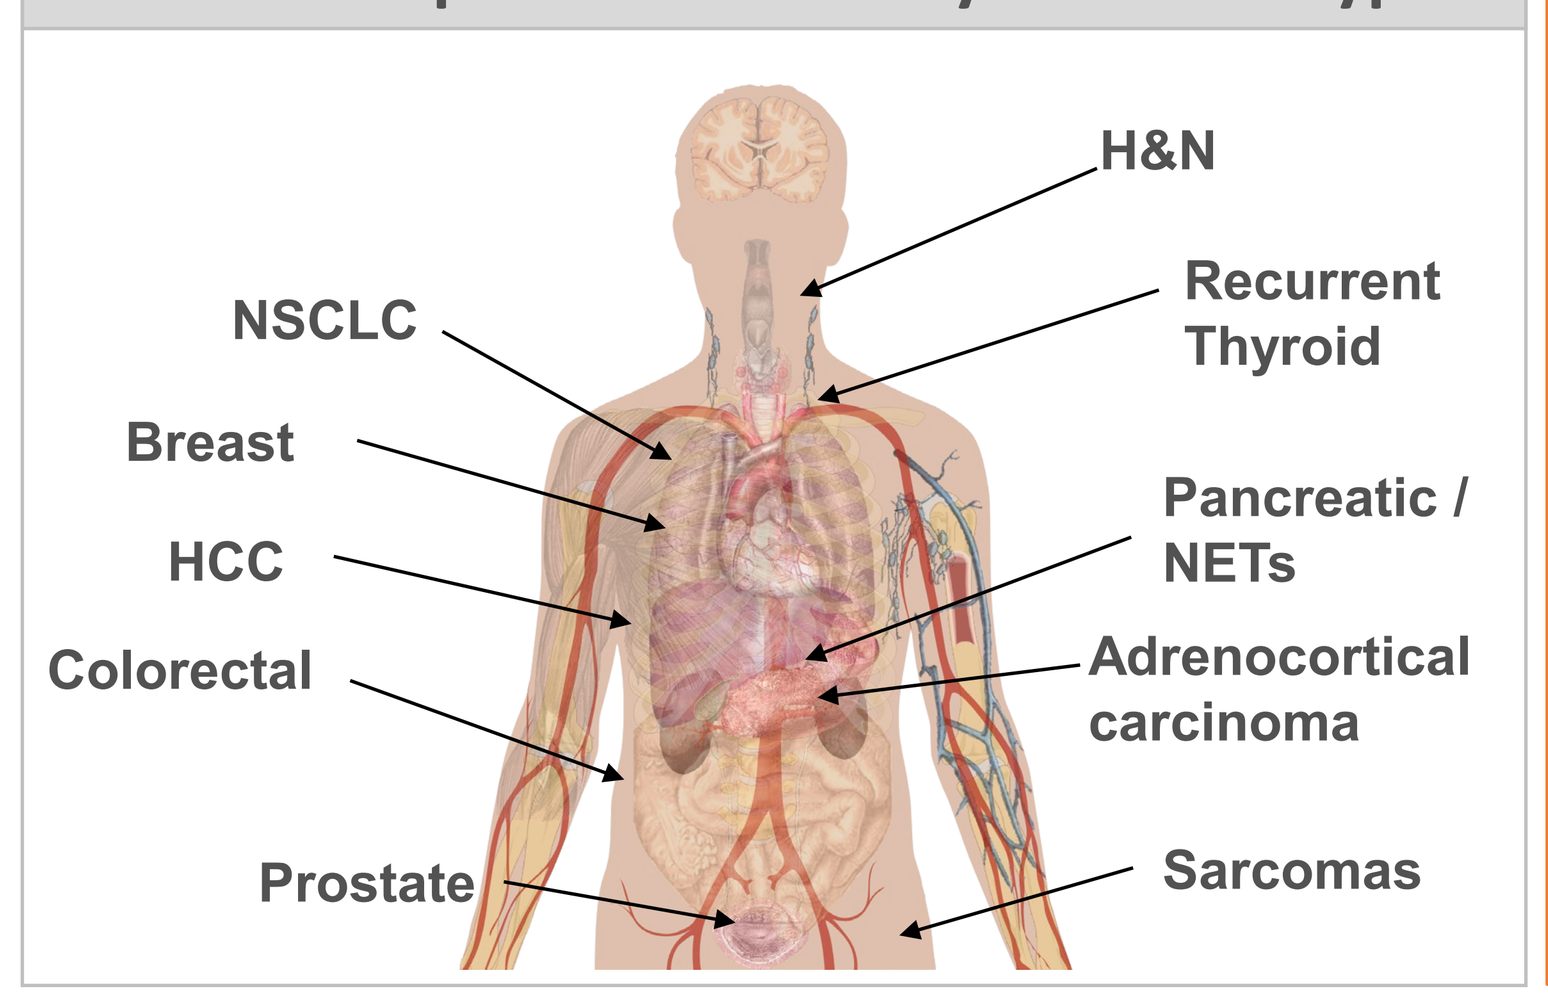
H&N
Recurrent Thyroid
NSCLC
Breast
Pancreatic / NETs
HCC
Adrenocortical carcinoma
Colorectal
Prostate
Sarcomas

Emw [CC BY-SA 3.0 (https://creativecommons.org/licenses/by-sa/3.0)]

IGF-1R has been implicated in:

- Increased cellular proliferation
- Metastatic potential
- Cell survival
- Chemotherapy and radiotherapy resistance

FPI-1434 Mechanism of Action

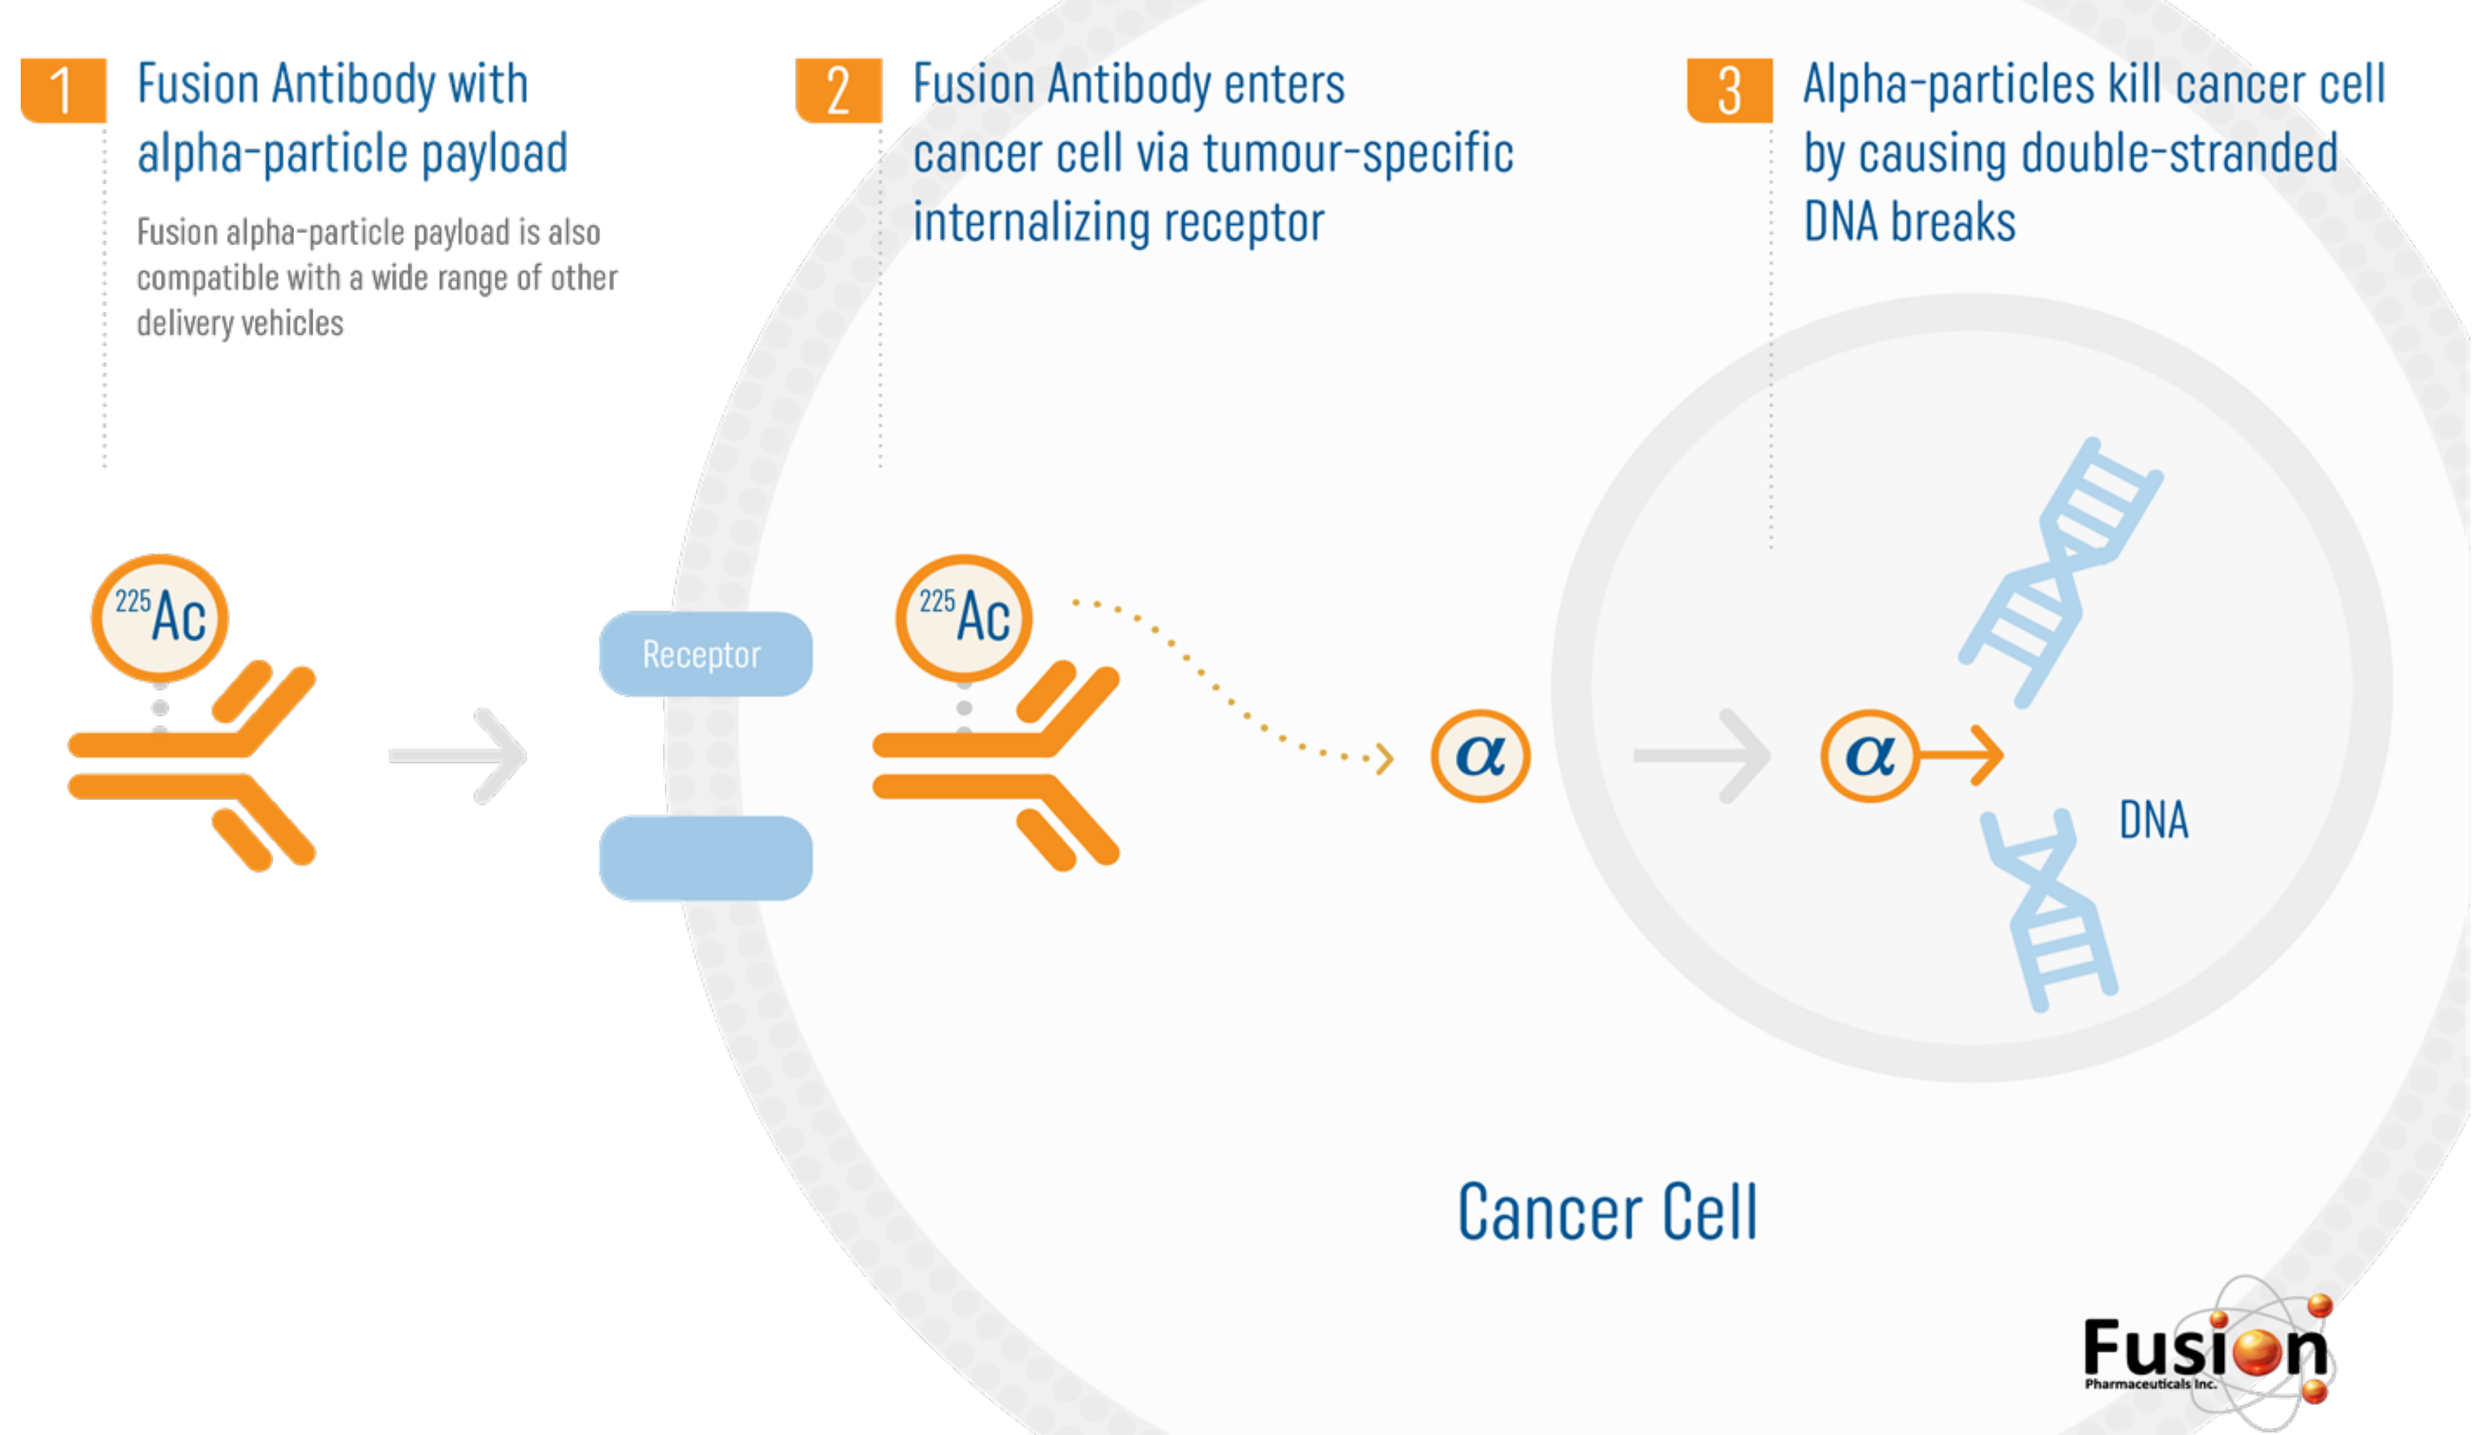
1 Fusion Antibody with alpha-particle payload
Fusion alpha-particle payload is also compatible with a wide range of other delivery vehicles
2 Fusion Antibody enters cancer cell via tumour-specific internalizing receptor
3 Alpha-particles kill cancer cell by causing double-stranded DNA breaks
225Ac
Receptor
α
DNA
Cancer Cell

FPI-1434 (Single dose, 400 nCi) Induced Double-Stranded DNA Breaks in Colo-205 Xenografts as Determined by γH2AX

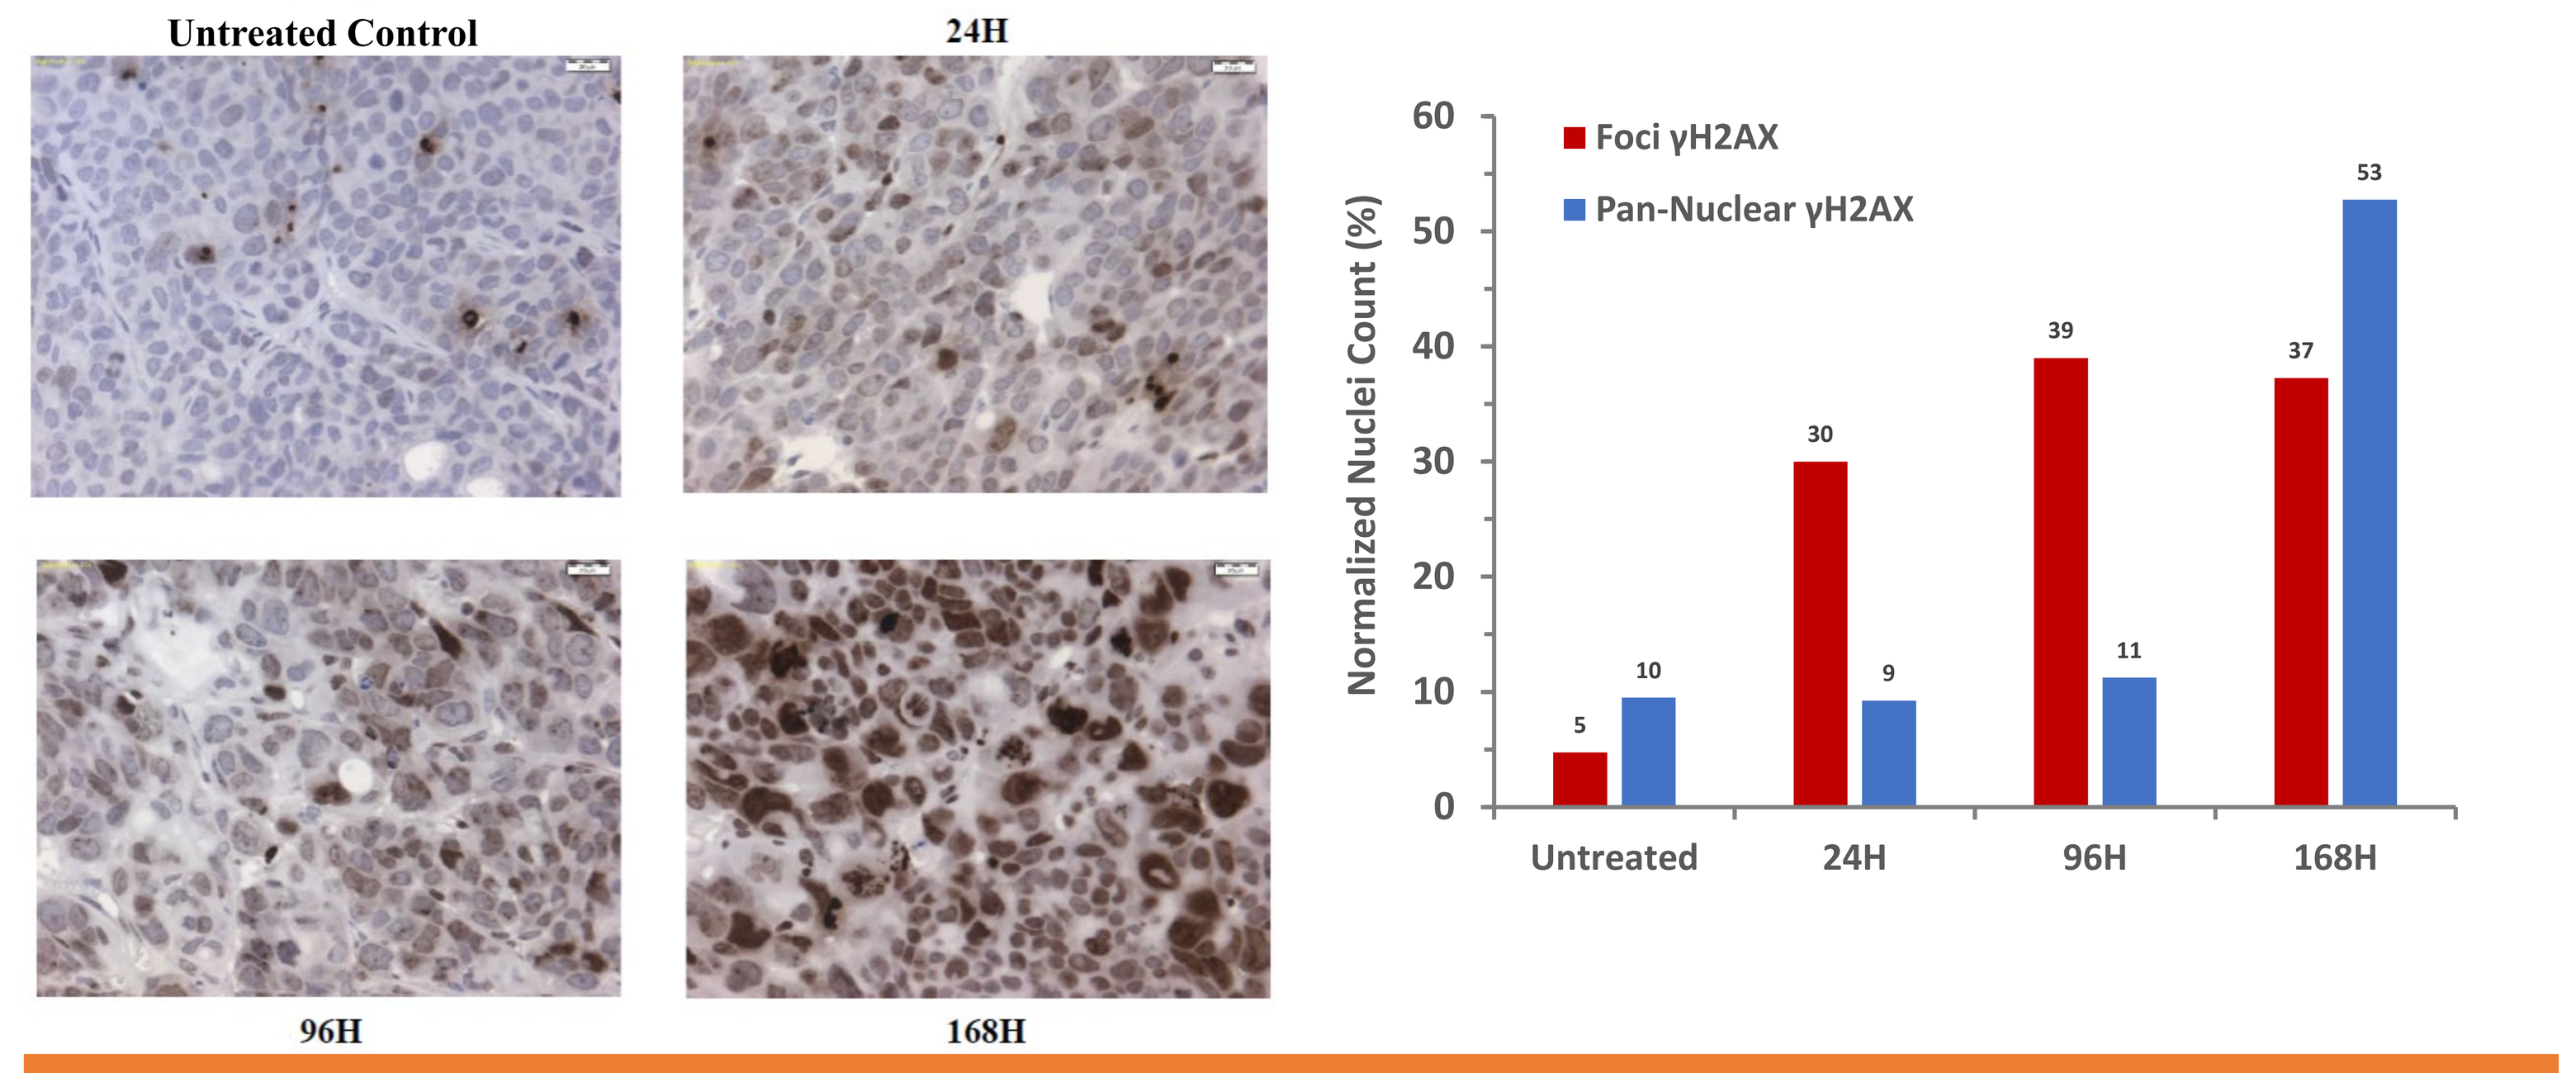
Untreated Control
24H
96H
168H
Foci γH2AX
Pan-Nuclear γH2AX
Normalized Nuclei Count (%)
Untreated 5 10
24H 30 9
96H 39 11
168H 37 53

PARP inhibition via olaparib co-therapy is a rational approach to potentiate tumor DSB formation driven by FPI-1434 treatment

- Delivery of alpha-particle radiation by FPI-1434 induced DSB in treated tumor xenografts (γH2Ax detected by IHC on fixed tumor sections)
 - DSB formation was time-dependent, dose dependent, and was consistent with kinetics of FPI-1434 tumor accumulation (data not shown)

FPI-1434 (Single dose, 400 nCi) Induced Apoptosis in Colo-205 Xenografts as Determined by Cleaved Caspase 3 (CC3)

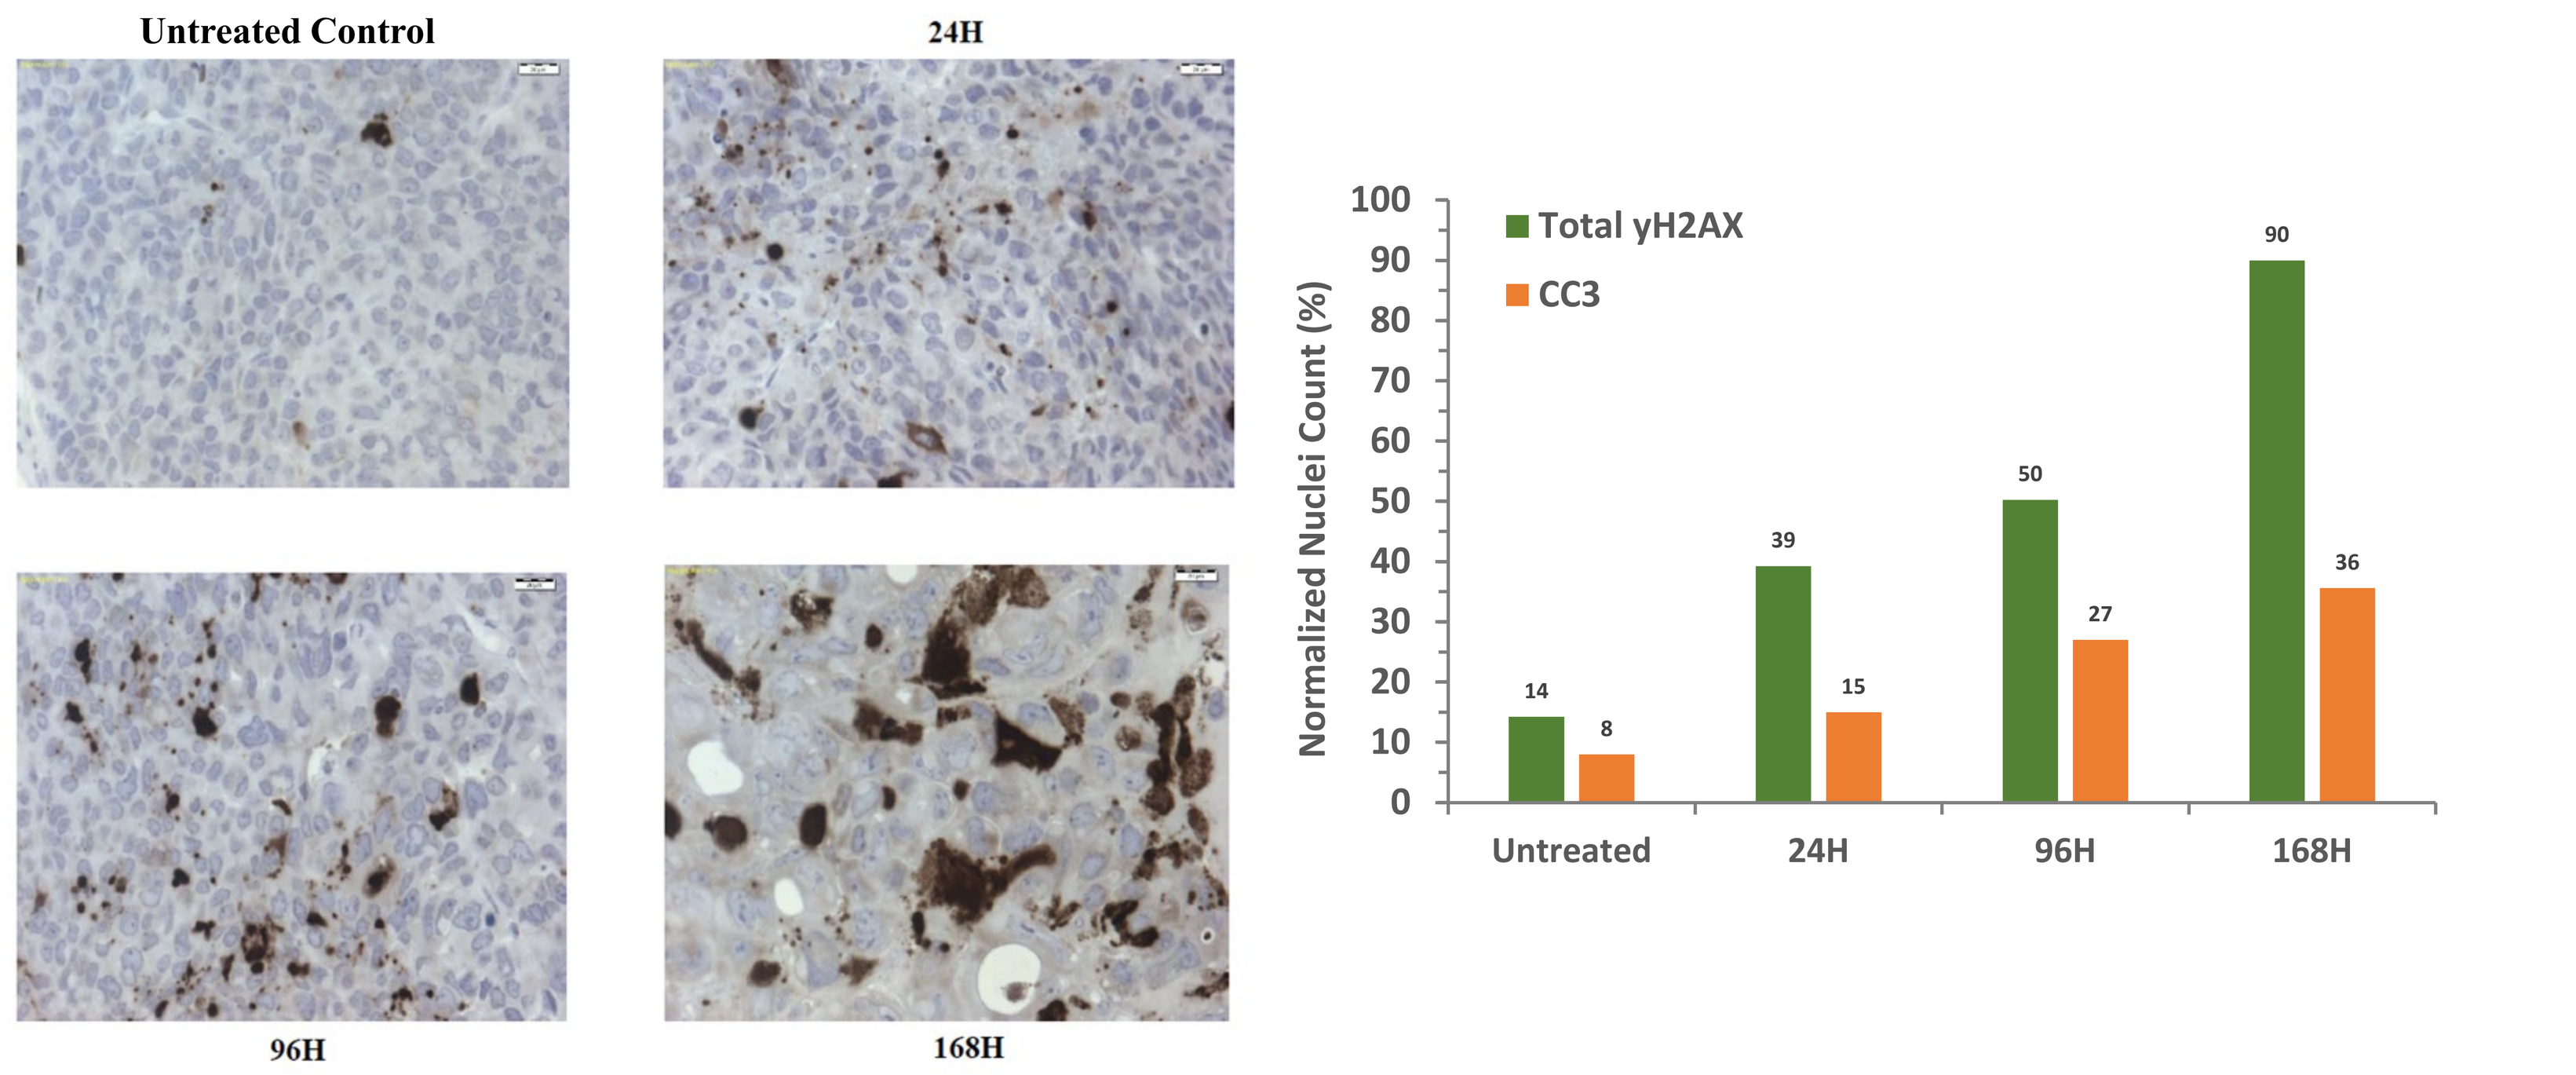
Untreated Control
24H
96H
168H
Total γH2AX
CC3
Normalized Nuclei Count (%)
Untreated 14 8
24H 39 15
96H 50 27
168H 90 36

- Delivery of alpha-particle radiation by FPI-1434 induced apoptosis in treated tumor xenografts (CC3 detected by IHC on fixed tumor sections)
 - Apoptosis was time dependent, following kinetics of tumor accumulation by FPI-1434 (data not shown), and was concomitant with induction of DSB
 - Apoptosis increased in conjunction with FPI-1434 dosage (data not shown)

FPI-1434 + Olaparib Therapy: Combined Efficacy at Low Single-Agent Doses (Colo-205, Colorectal Cancer Xenograft Model)

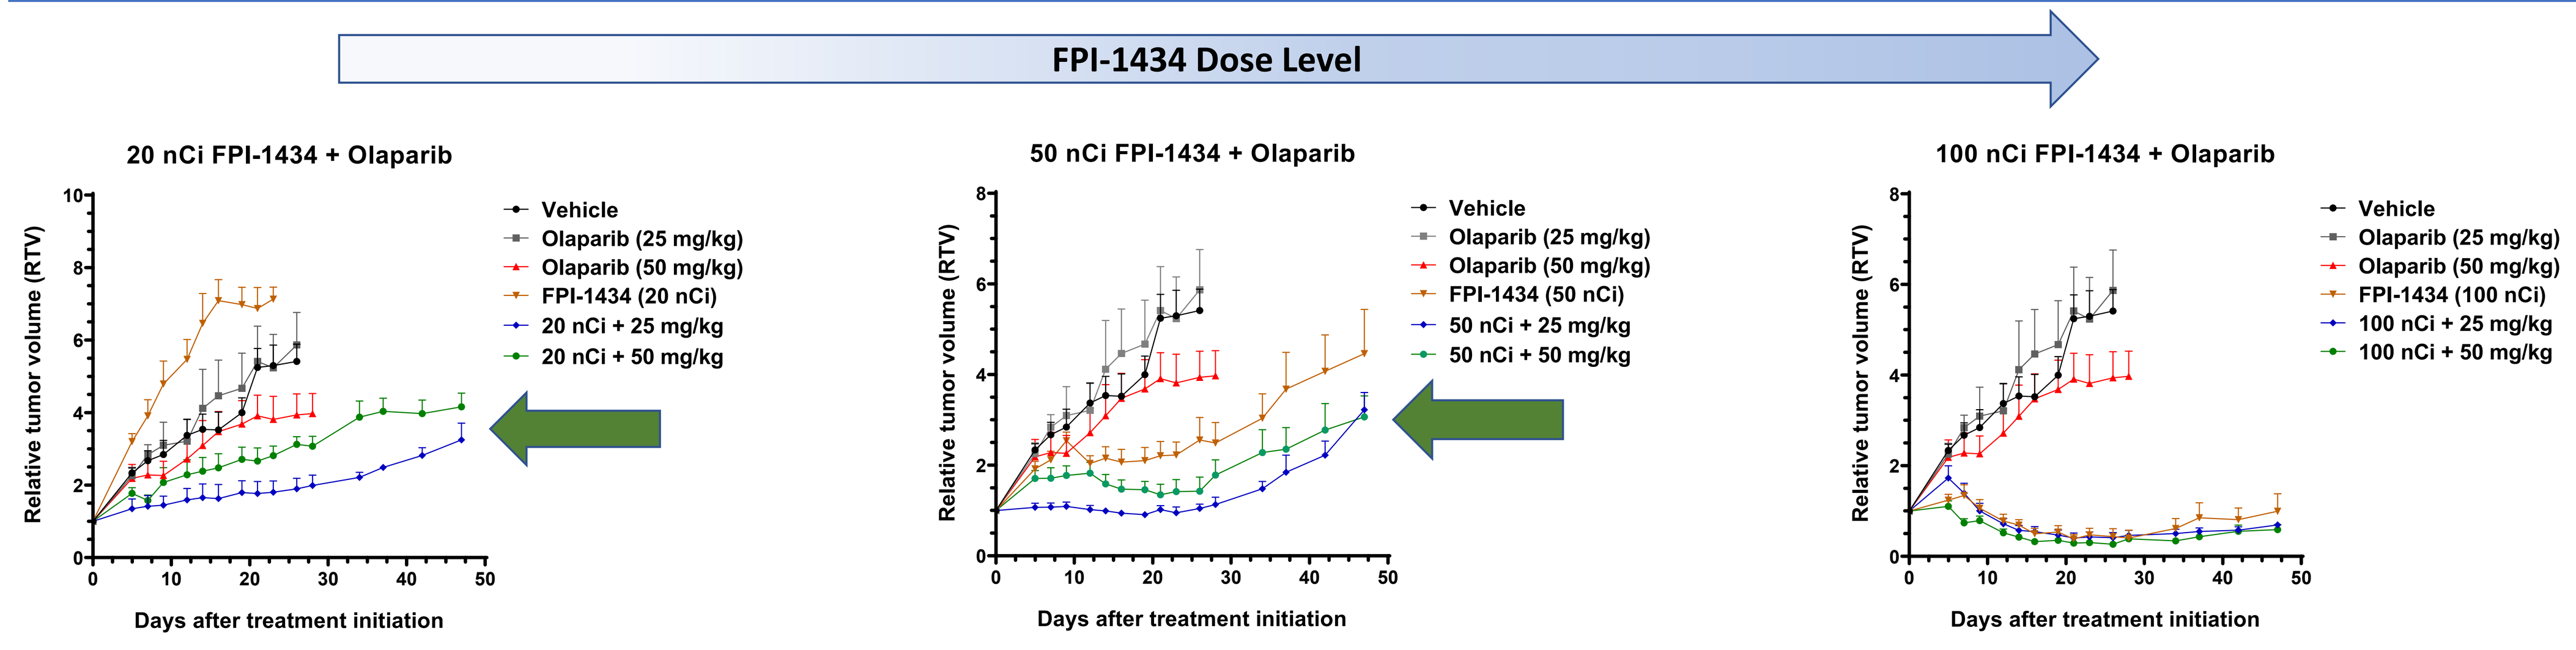
FPI-1434 Dose Level
20 nCi FPI-1434 + Olaparib
50 nCi FPI-1434 + Olaparib
100 nCi FPI-1434 + Olaparib
Relative tumor volume (RTV)
Days after treatment initiation
Vehicle
Olaparib (25 mg/kg)
Olaparib (50 mg/kg)
FPI-1434 (20 nCi)
20 nCi + 25 mg/kg
20 nCi + 50 mg/kg
FPI-1434 (50 nCi)
50 nCi + 25 mg/kg
50 nCi + 50 mg/kg
FPI-1434 (100 nCi)
100 nCi + 25 mg/kg
100 nCi + 50 mg/kg

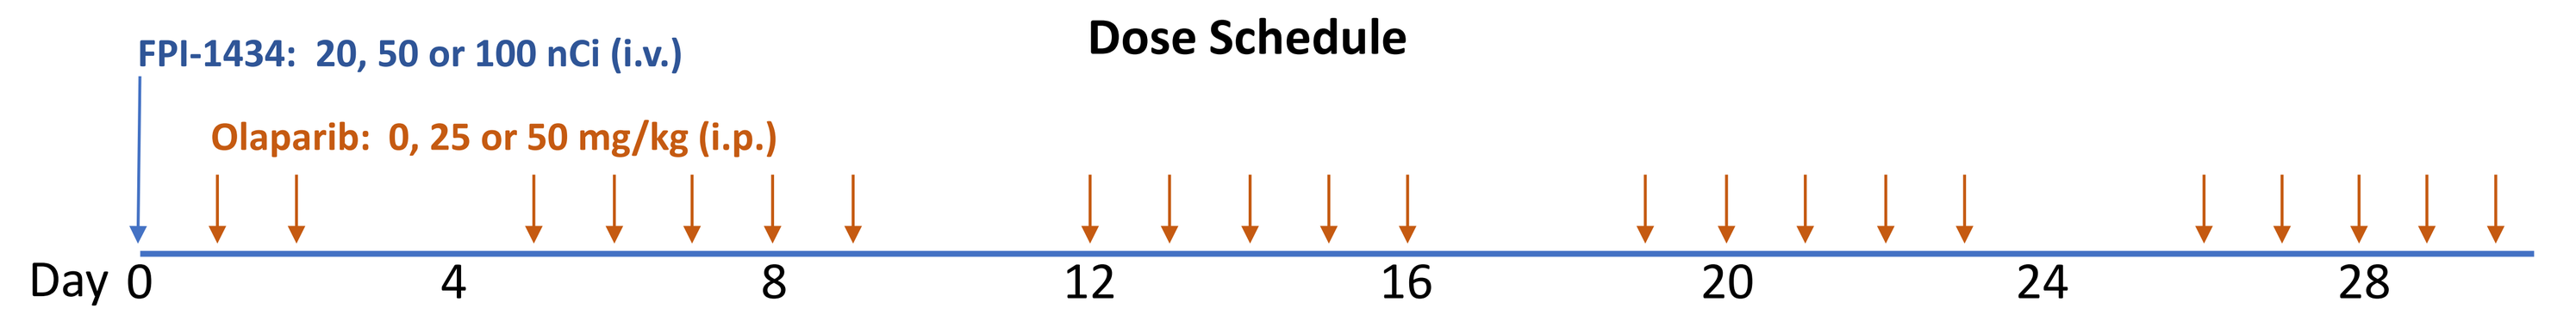
Dose Schedule
FPI-1434: 20, 50 or 100 nCi (i.v.)
Olaparib: 0, 25 or 50 mg/kg (i.p.)
Day 0
4
8
12
16
20
24
28

FPI-1434 + Olaparib Therapy: Combined Efficacy at Low Single-Agent Doses (A549, NSCLC Xenograft Model)

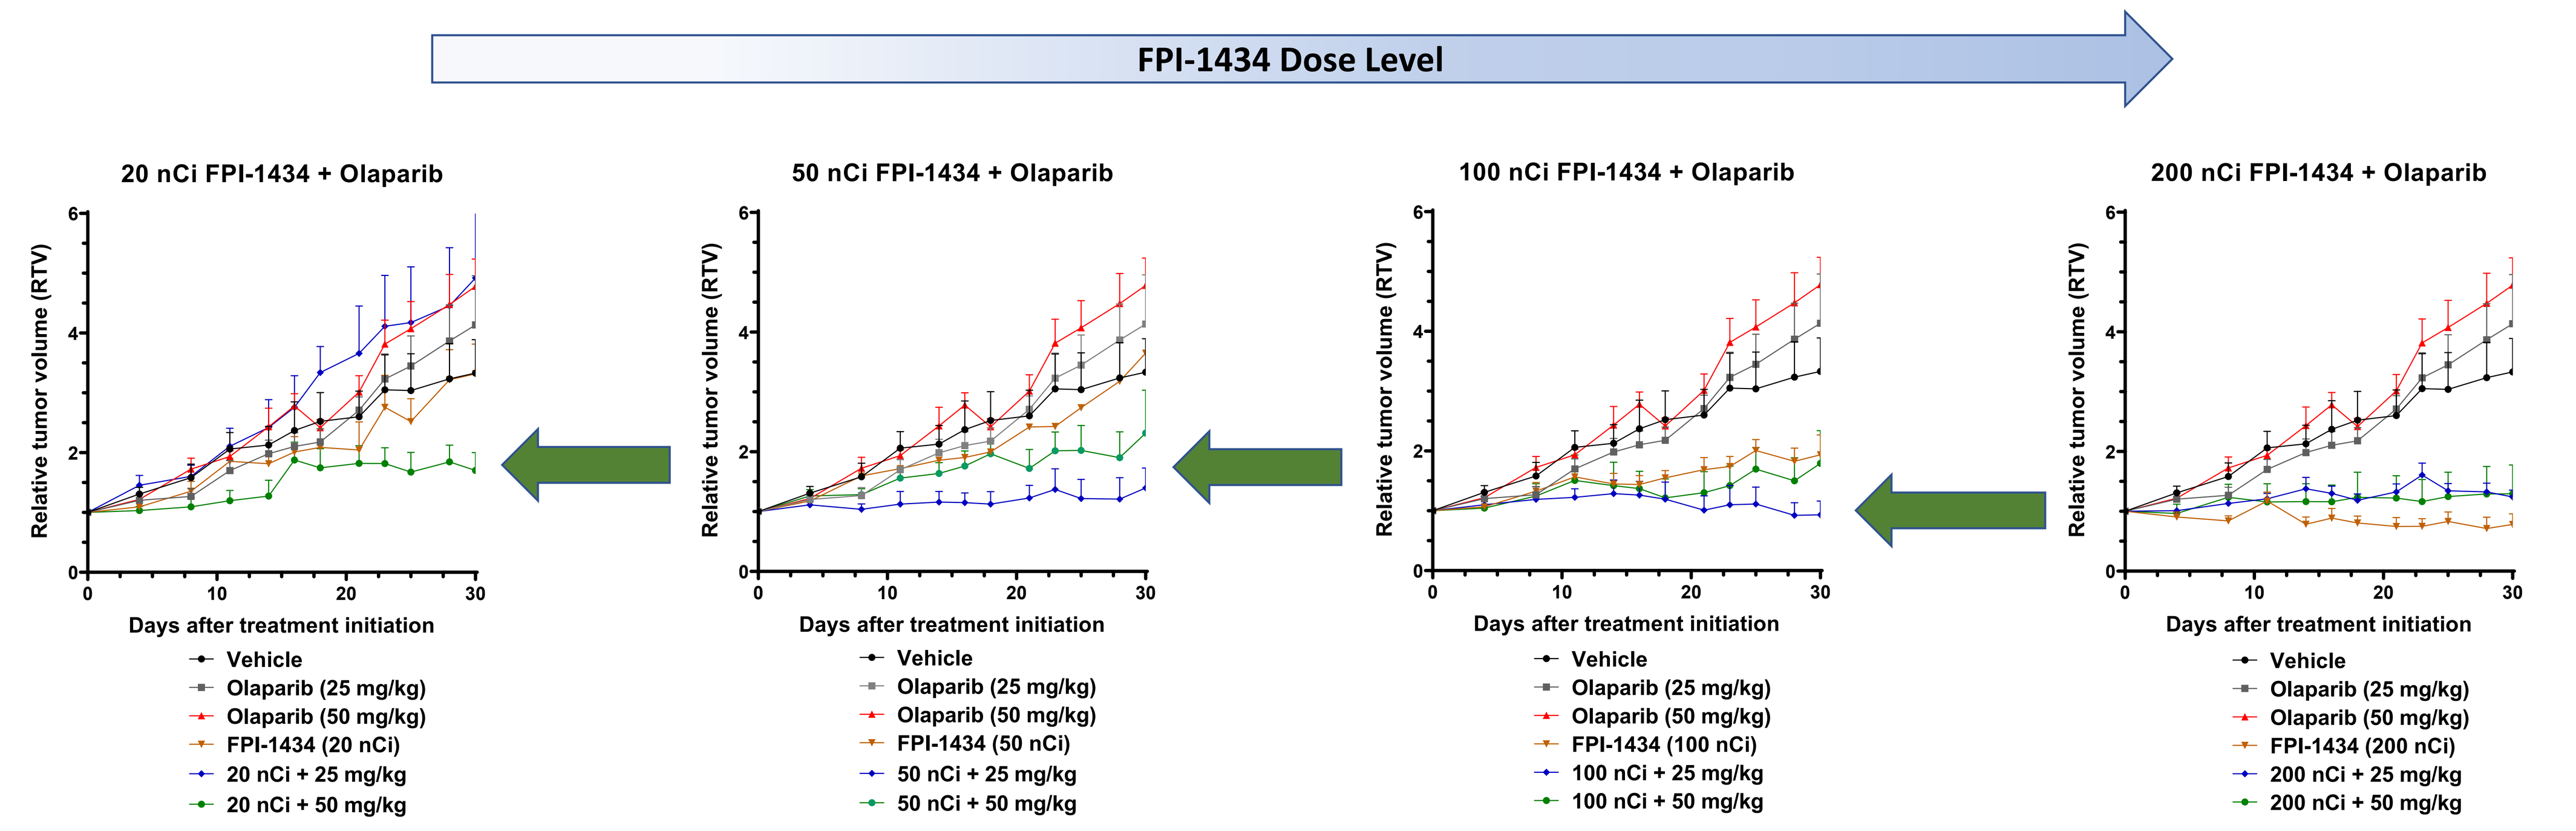
FPI-1434 Dose Level
20 nCi FPI-1434 + Olaparib
50 nCi FPI-1434 + Olaparib
100 nCi FPI-1434 + Olaparib
200 nCi FPI-1434 + Olaparib
Relative tumor volume (RTV)
Days after treatment initiation
Vehicle
Olaparib (25 mg/kg)
Olaparib (50 mg/kg)
FPI-1434 (20 nCi)
20 nCi + 25 mg/kg
20 nCi + 50 mg/kg
FPI-1434 (50 nCi)
50 nCi + 25 mg/kg
50 nCi + 50 mg/kg
FPI-1434 (100 nCi)
100 nCi + 25 mg/kg
100 nCi + 50 mg/kg
FPI-1434 (200 nCi)
200 nCi + 25 mg/kg
200 nCi + 50 mg/kg

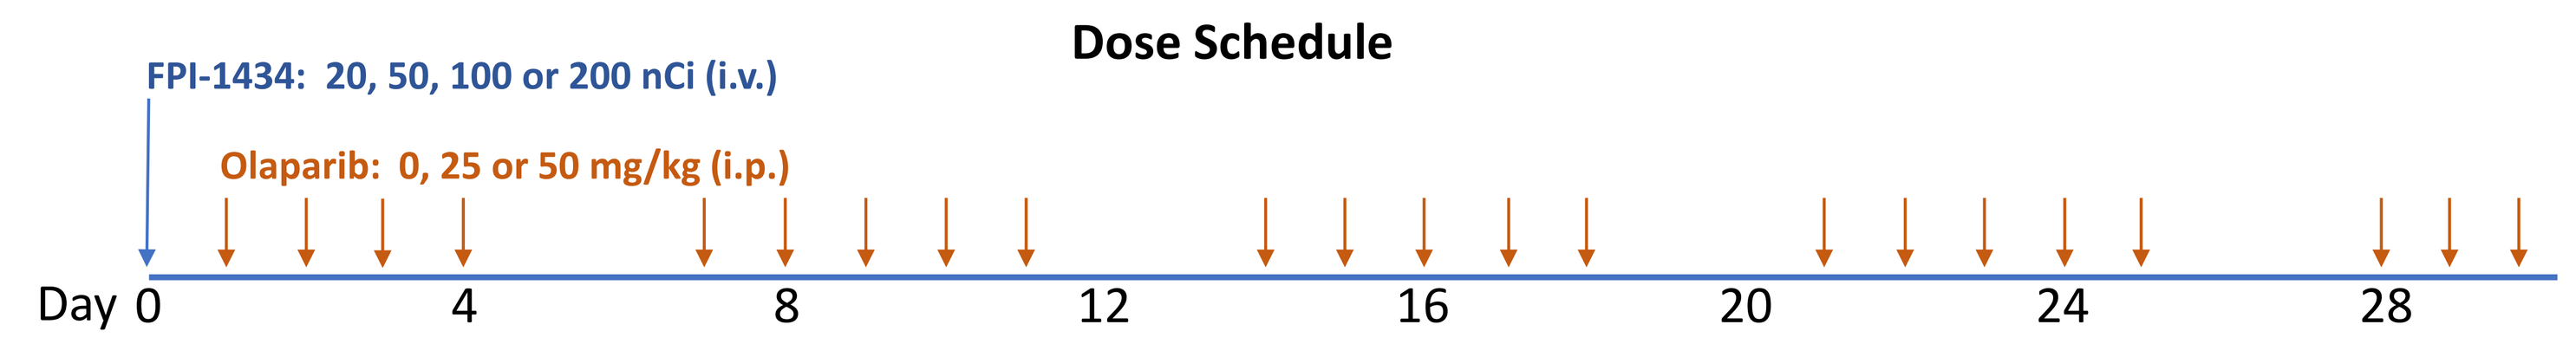
Dose Schedule
FPI-1434: 20, 50, 100 or 200 nCi (i.v.)
Olaparib: 0, 25 or 50 mg/kg (i.p.)
Day 0
4
8
12
16
20
24
28

Summary and Conclusions

- Delivery of alpha-particle radiation by FPI-1434 resulted in dose-dependent regression and induced double-stranded DNA breaks and apoptosis in tumor xenografts
- Combination of FPI-1434 and olaparib resulted in efficacy in the Colo-205 model and in the radioresistant A549 model
 - Olaparib treatment alone was not efficacious; models tested lack mutations in DNA repair pathways required for sensitivity to PARPi monotherapy
 - Strongest combination effects occurred at doses where FPI-1434 was not effective as a single agent
 - Colo-205: 20 nCi FPI-1434 + 25 mg/kg olaparib
 - A549: 50 nCi FPI-1434 + 25 mg/kg olaparib
 - FPI-1434 effect dominated at the highest single agent doses suggesting it was the therapeutic driver of the combination
- Olaparib co-dosing enhanced FPI-1434 therapeutic efficacy which supports consideration of this combination for clinical use
 - PARP inhibition while using FPI-1434 as the therapeutic driver of DSB formation can be applied to multiple cancer types where predisposing mutations in DNA repair are lacking
 - PARP inhibition may potentiate efficacy at lower clinical doses of FPI-1434