Abstract TPS3152

2019 ASCO ANNUAL MEETING

A Phase I Study of [225Ac]-FPI-1434 Radioimmunotherapy in Patients with IGF-1R Expressing Solid Tumors

Rosalyn A. Juergens[1], Katherine A. Zukotynski[2], Daniel Juneau[3], Ryan Simms[4], John Forbes[4], Eric S. Burak[4], John Valliant[4], Lily Krnezich[2], Lauren Stafford[4], Thomas Armor[4], Istvan Molnar[4], Fred Saad[3]

[1]Juravinski Cancer Centre, McMaster University, Hamilton, ON; [2]Nuclear Medicine and Radiology, McMaster University, Hamilton, ON; [3]Centre Hospitalier de l'Université de Montréal, Université de Montréal, Montreal, QC; [4]Fusion Pharmaceuticals, Hamilton, ON and Boston, MA

Background

Type I insulin-like growth factor receptor (IGF-1R) is a transmembrane protein which is overexpressed in solid tumors including non–small cell lung, prostate, sarcomas, and breast cancers.

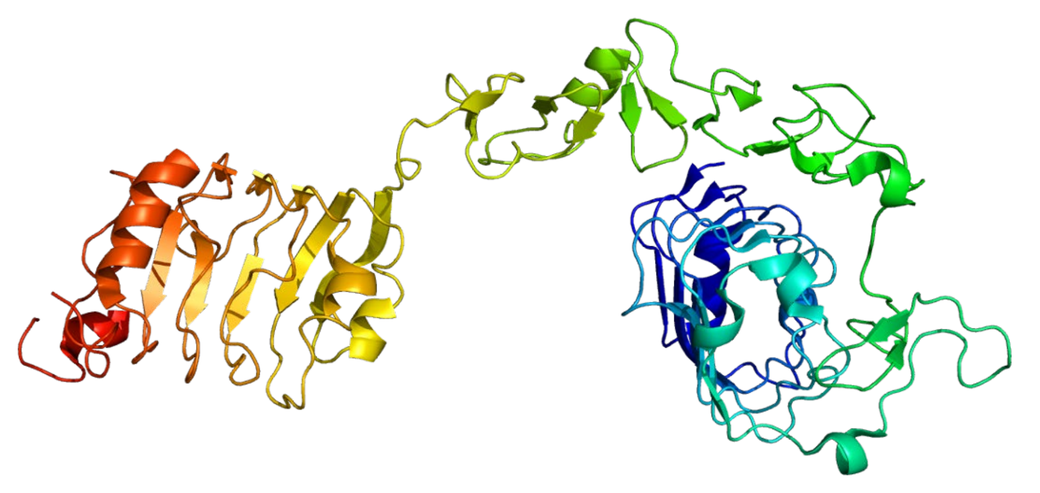

IGF-1R

IGF-1R has been implicated in:
- Increased cellular proliferation
- Metastatic potential
- Cell survival
- Chemotherapy and radiotherapy resistance

IGF-1R is expressed on nearly all tumor types

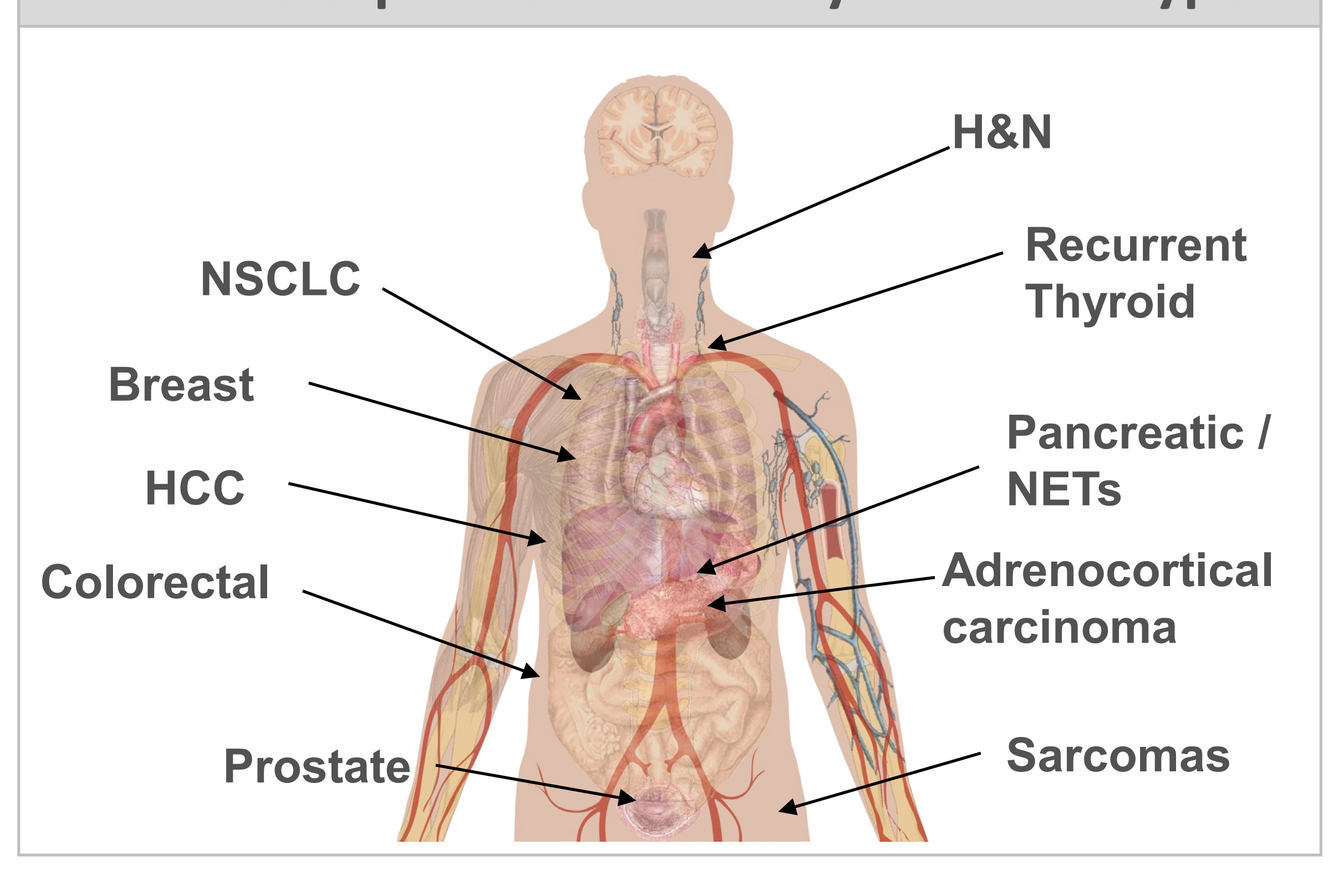
H&N
NSCLC
Recurrent Thyroid
Breast
HCC
Pancreatic / NETs
Colorectal
Adrenocortical carcinoma
Prostate
Sarcomas

Emw [CC BY-SA 3.0 (https://creativecommons.org/licenses/by-sa/3.0)]

[225Ac]-FPI-1434 is a radioimmunoconjugate consisting of a humanized monoclonal antibody (AVE1642) that binds to the external domain of IGF-1R, a proprietary bifunctional chelate, and the alpha-emitting radionuclide actinium-225 (Ac-225). Internalization of the radioimmunoconjugate and decay of Ac-225 causes tumor cell death primarily through double stranded DNA breaks. The indium-111 analog, [111In]-FPI-1547, with the identical antibody and bifunctional chelate is used for patient selection and quantification of IGF-1R expressing targets prior to therapy.

Imaging | Therapy

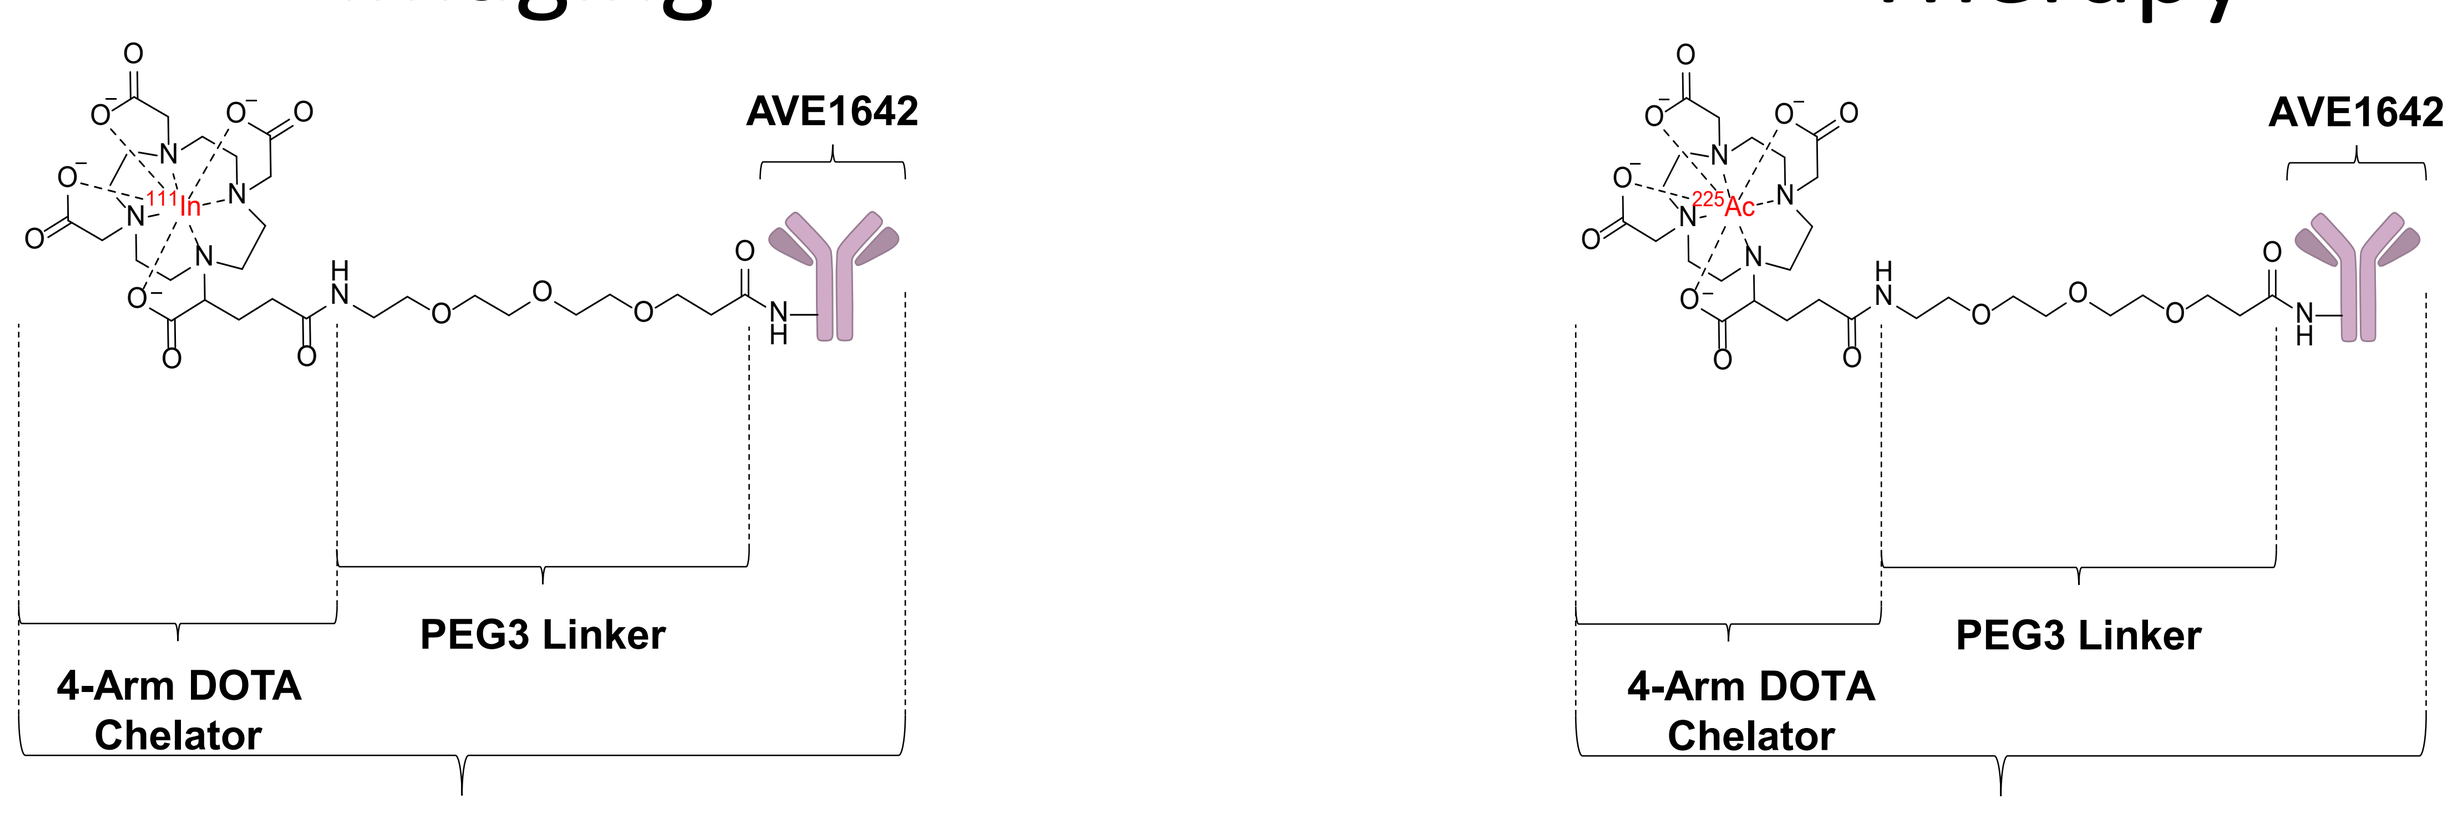
AVE1642
PEG3 Linker
4-Arm DOTA Chelator
AVE1642
PEG3 Linker
4-Arm DOTA Chelator

[111In]-FPI-1547 | **[225Ac]-FPI-1434**

Figure 1. Mechanism of Action

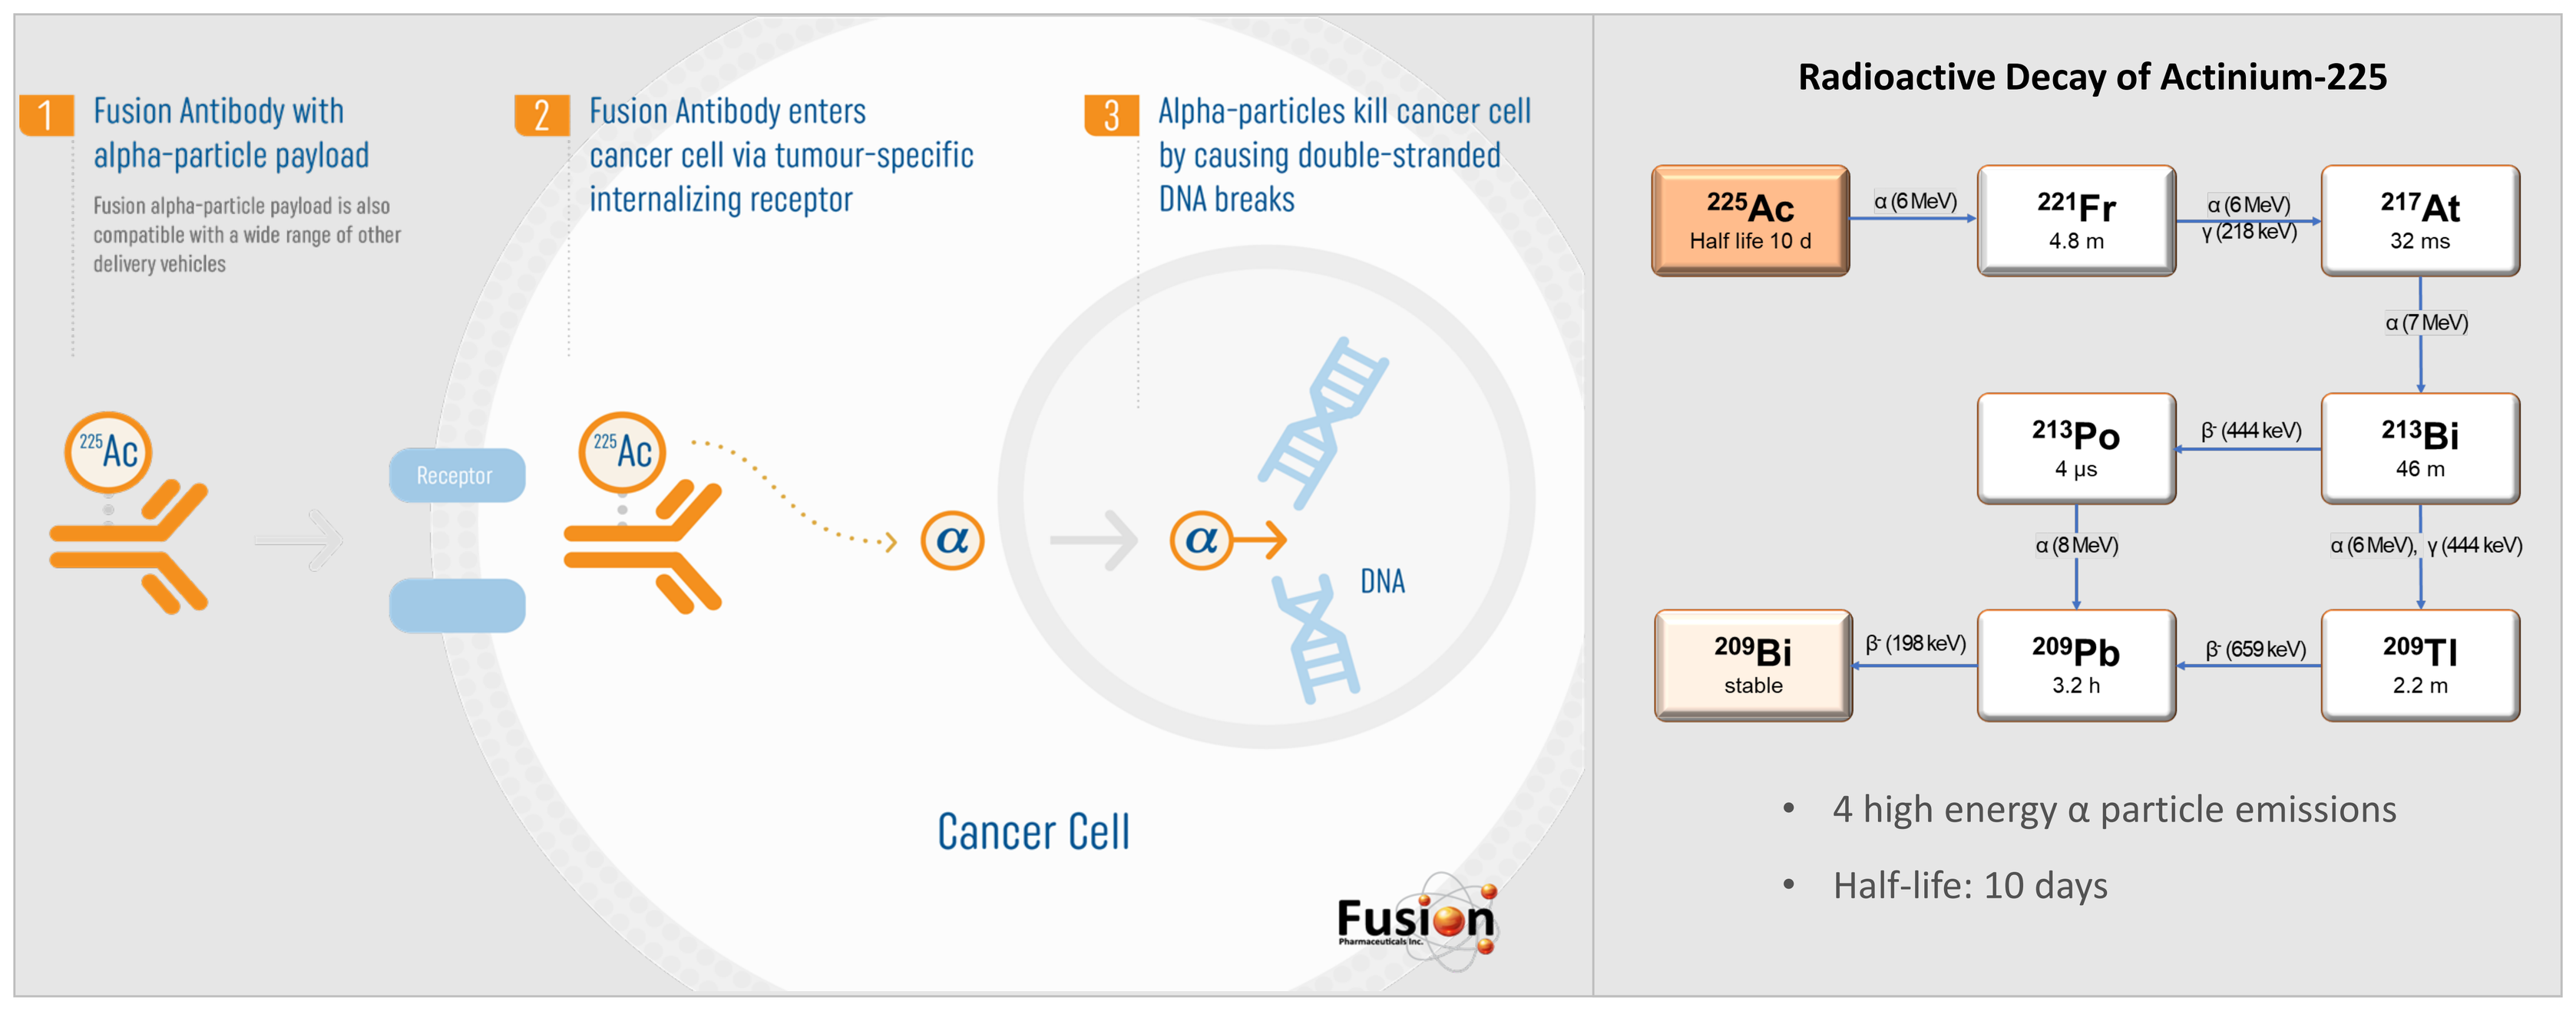
1 Fusion Antibody with alpha-particle payload
Fusion alpha-particle payload is also compatible with a wide range of other delivery vehicles
2 Fusion Antibody enters cancer cell via tumour-specific internalizing receptor
3 Alpha-particles kill cancer cell by causing double-stranded DNA breaks
225Ac
Receptor
DNA
Cancer Cell
Fusion
Radioactive Decay of Actinium-225
225Ac Half life 10 d
α (6 MeV)
221Fr 4.8 m
α (6 MeV) γ (218 keV)
217At 32 ms
α (7 MeV)
213Po 4 µs
β⁻ (444 keV)
213Bi 46 m
α (8 MeV)
α (6 MeV), γ (444 keV)
209Bi stable
β⁻ (198 keV)
209Pb 3.2 h
β⁻ (659 keV)
209Tl 2.2 m
4 high energy α particle emissions
Half-life: 10 days

Based on anti-tumor activity of [225Ac]-FPI-1434 in non-clinical models, favorable toxicology studies in cynomolgus monkeys, and prior human experience with the unconjugated antibody, the first-in-human trial was initiated.

Figure 2. Single Dose Therapeutic Efficacy in a Colorectal Cancer Tumor Xenograft Model (Colo-205)

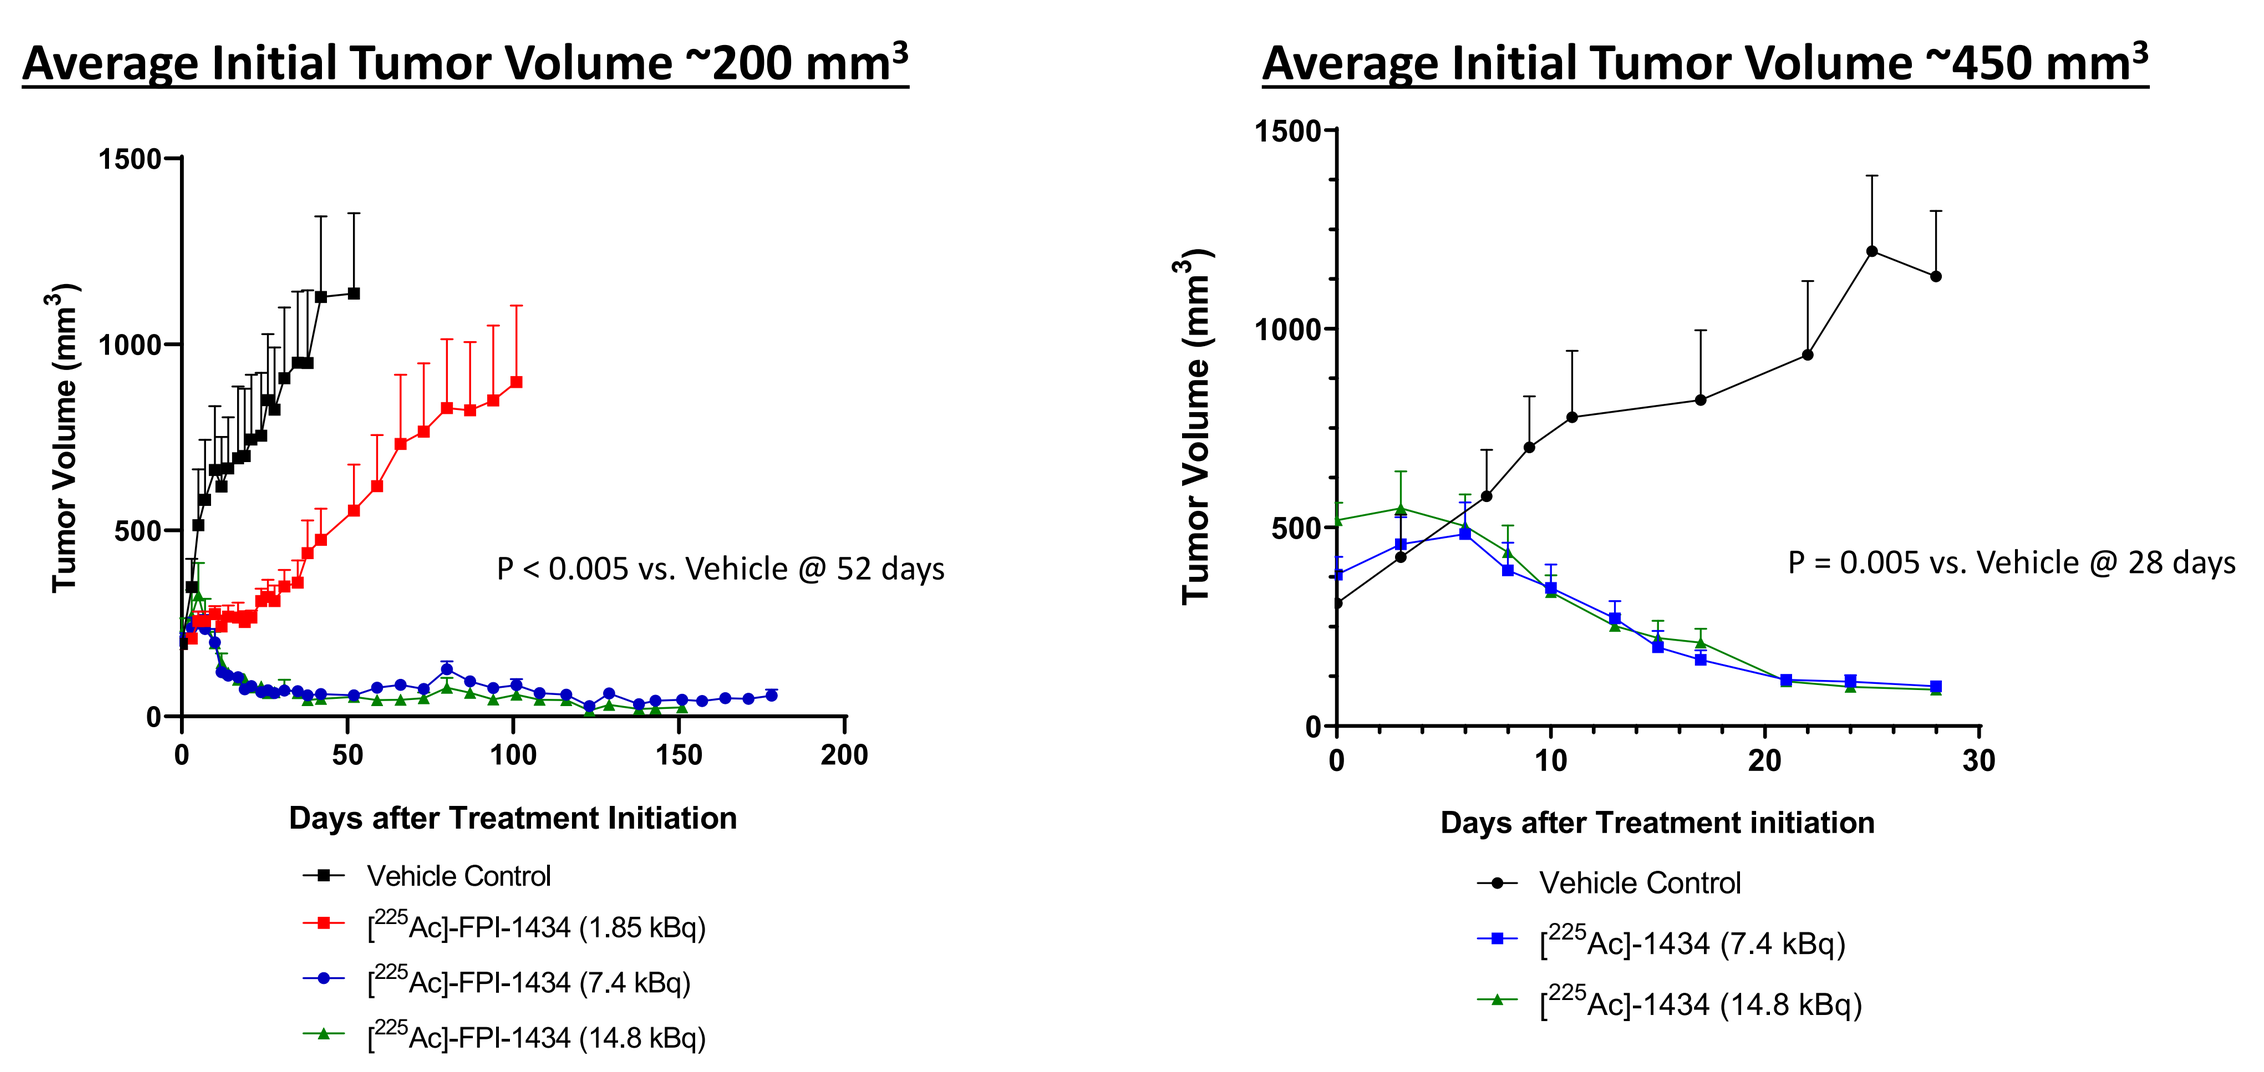
Average Initial Tumor Volume ~200 mm³
Tumor Volume (mm³)
P < 0.005 vs. Vehicle @ 52 days
Days after Treatment Initiation
Vehicle Control
[225Ac]-FPI-1434 (1.85 kBq)
[225Ac]-FPI-1434 (7.4 kBq)
[225Ac]-FPI-1434 (14.8 kBq)
Average Initial Tumor Volume ~450 mm³
Tumor Volume (mm³)
P = 0.005 vs. Vehicle @ 28 days
Days after Treatment initiation
Vehicle Control
[225Ac]-1434 (7.4 kBq)
[225Ac]-1434 (14.8 kBq)

Objectives

Primary
- Evaluate the safety and tolerability of [111In]-FPI-1547 Injection and [225Ac]-FPI-1434 Injection in patients with advanced refractory solid tumors
- Determine the maximum tolerated dose of a single [225Ac]-FPI-1434 Injection

Secondary
- Obtain preliminary data on tumor uptake of [111In]-FPI-1547 Injection in cancer patients
- Estimate the dosimetry for [225Ac]-FPI-1434 Injection (whole body and individual regions)
- Determine the pharmacokinetics of [111In]-FPI-1547 Injection and [225Ac]-FPI-1434 Injection
- Describe efficacy of [225Ac]-FPI-1434 Injection
- Assess the effect of [225Ac]-FPI-1434 on QTc prolongation
- Assess changes in human growth hormone (hGh), insulin-like growth factor (IGF)-1 and insulin-like growth binding protein 3 (IGFBP-3) following [111In]-FPI-1547 and [225Ac]-FPI-1434 injections

Key Eligibility Criteria

Inclusion:
- Age ≥18 years old
- Pathologically documented, definitively diagnosed, advanced solid tumor that is refractory to all standard treatment, for which no standard treatment is available
- At least 1 measurable lesion (≥20 mm in largest diameter [≥20 mm in shortest diameter if lymph node])
- Eastern Cooperative Oncology Group (ECOG) Performance status of 0 or 1
- Life expectancy equal to or greater than 3 months as judged by the treating physician
- Available tumor tissue (either archival or fresh biopsy) for IGF-1R immunohistochemistry
- Adequate bone marrow reserves without the use of hematopoietic growth factors, red cell or platelet transfusion
- Adequate renal function as evidenced by a creatinine clearance ≥60 mL/min using the Cockcroft-Gault Equation
- Adequate hepatic function
- Sufficient target expression in at least one tumor lesion following the [111In]-FPI-1547 Injection
- Dosimetry estimate of the planned [225Ac]-FPI-1434 Injection does not exceed predetermined limits for kidney, liver and lungs

Exclusion:
- Systemic therapeutic radiopharmaceutical within 6 months prior to enrollment
- Contraindications to or inability to perform the imaging procedures required in this study
- Uncontrolled brain metastasis, including but not limited to need for treatment with steroids, surgery or radiation therapy
- Anticancer therapy (including investigations agents) or external beam radiation therapy within 14 days of the dosing of [111In]-FPI-1547 (6 weeks for mitomycin-C)
- Prior organ transplantation, including stem cell transplantation.
- Any prior treatment with nitrosoureas and actinomycin-D
- Clinical relevant proteinuria
- Known or suspect allergies or contraindications to the Investigational Products or any component of the investigational drug formulation
- Uncontrolled intercurrent illness including, but not limited to, ongoing or active infection, symptomatic congestive heart failure, unstable angina pectoris, cardiac arrhythmia, diabetes, or psychiatric illness/social situations that would limit compliance with study requirements
- Received > 20 Gy prior radiation to large areas of the bone marrow

Study Design

- Study FPX-01-01 (NCT03746431)
- Open-label, multi-center phase I study of a single injection of Ac-225 radioimmunotherapy conjugate (option for a second dose if clinical benefit)
- Modified 3+3 dose-escalation (5 dose cohorts)
- Followed by an 8-week DLT evaluation period.

Dose Cohort	Administered Dosage of [225Ac]-FPI-1434
1	10 kBq/kg body weight (0.27 µCi/kg body weight)
2	20 kBq/kg body weight (0.54 µCi/kg body weight)
3	40 kBq/kg body weight (1.1 µCi/kg body weight)
4	80 kBq/kg body weight (2.2 µCi/kg body weight)
5	120 kBq/kg body weight (3.2 µCi/kg body weight)

GENERAL SCREENING

- Informed consent
- Safety laboratory assessments
- Tumor biopsy if needed
- Baseline tumor imaging
- Patient meets general eligibility criteria

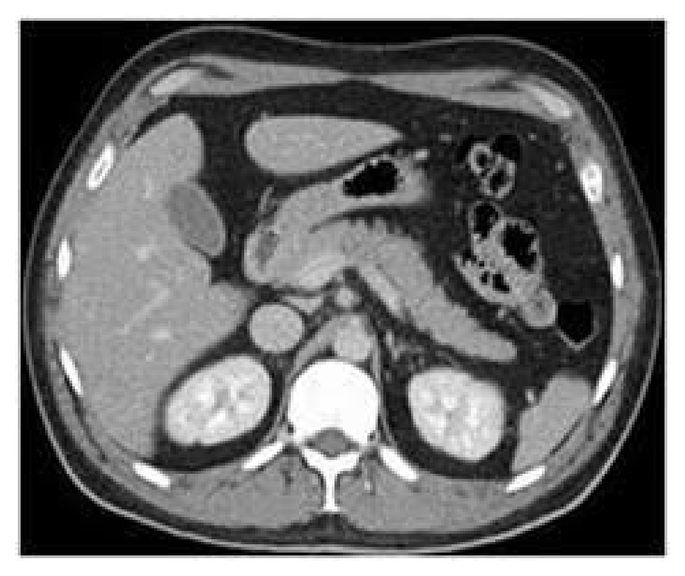

IMAGING SCREENING
[111In]-FPI-1547 Injection

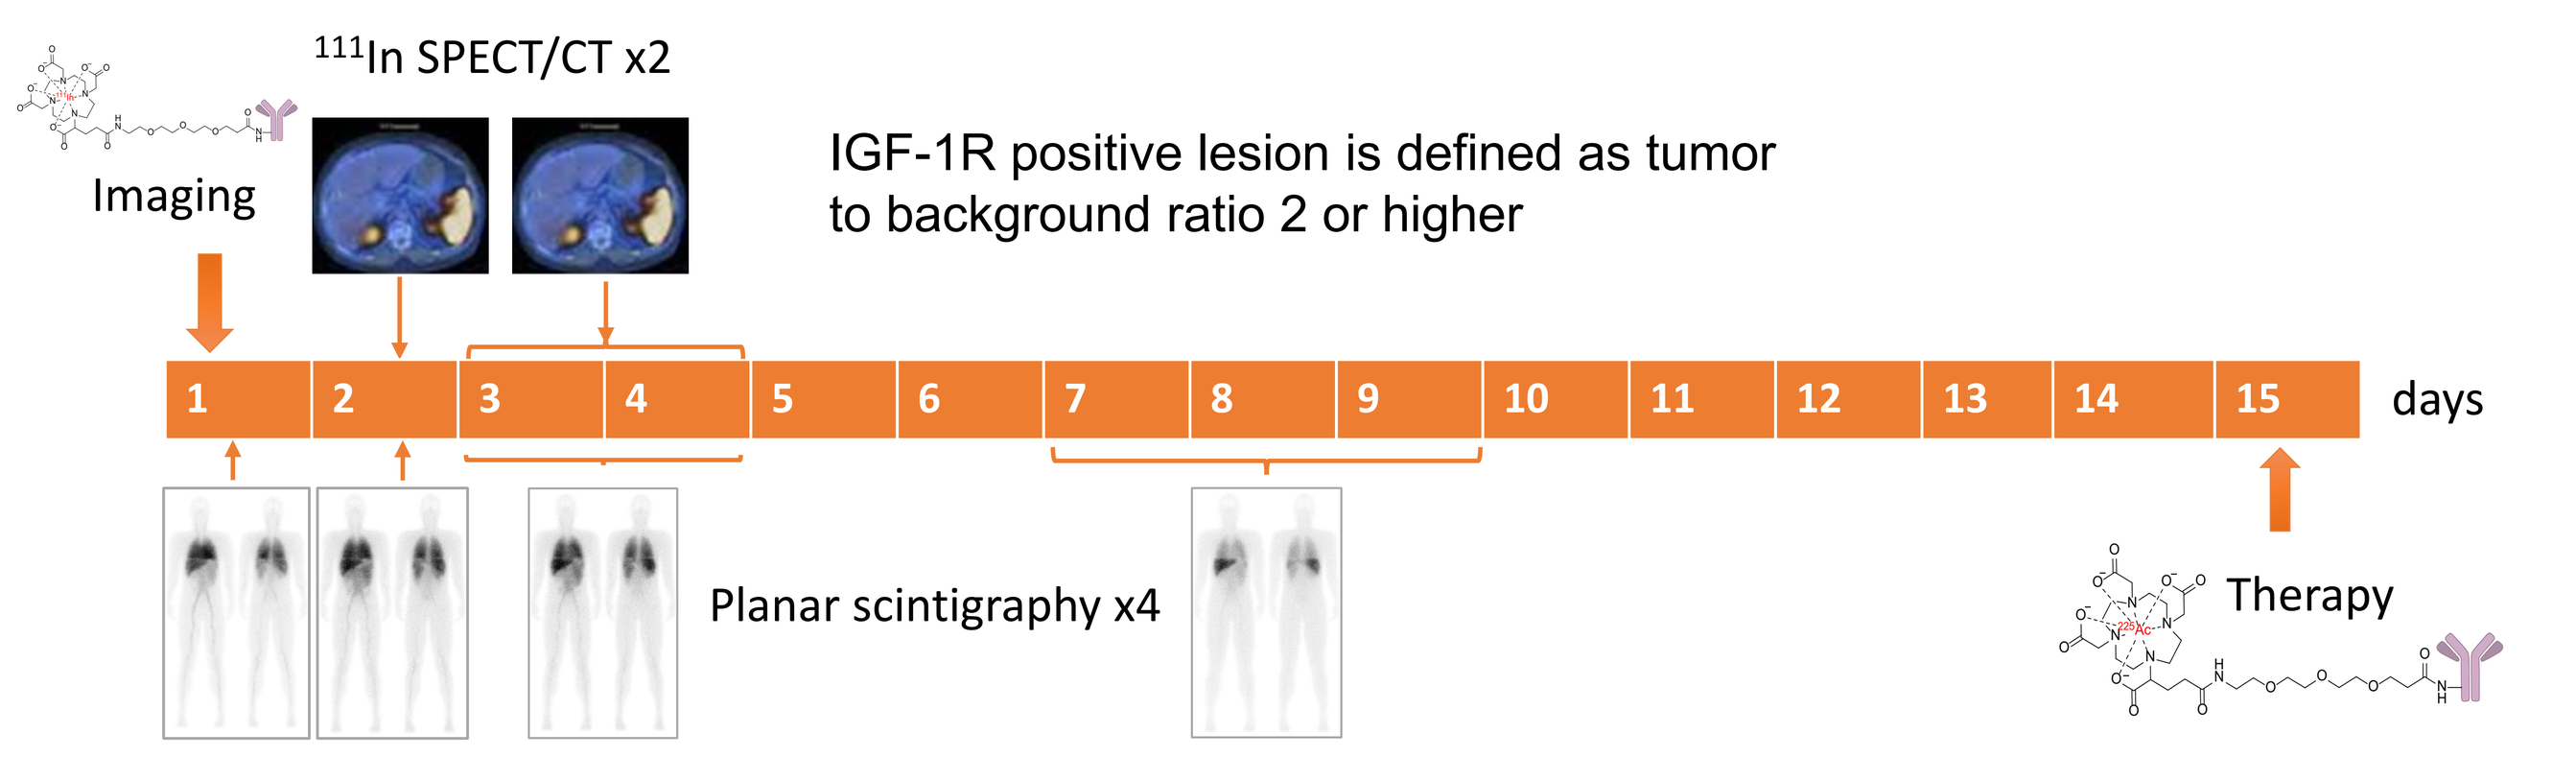
Imaging
111In SPECT/CT x2
IGF-1R positive lesion is defined as tumor to background ratio 2 or higher
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
days
Planar scintigraphy x4
Therapy

Infuse only if IGF-1R positive on at least one lesion and dosimetry estimate for 225Ac is acceptable

TREATMENT PERIOD
[225Ac]-FPI-1434 Injection

Weekly safety assessments for DLT period (56 days)

FOLLOW-UP PERIOD

Until tumor progression / initiation of a new systemic anticancer therapy, or 6 months post [225Ac]-FPI-1434 Injection, whichever is later:

- Tumor assessments every 8 weeks for 6 months then every 3 months until progression or until initiation of new systemic anticancer therapy, whichever is sooner
- Safety assessments at 3, 4, 5, and 6 mos. dependent on patient's clinical condition

Study Status

Currently enrolling patients at the following locations:
- Juravinski Cancer Center (Hamilton Health Sciences) – Hamilton, Ontario
- Quebec Hospital/Laval – Quebec City, Quebec
- Universite de Montreal (CHUM) – Montréal, Québec

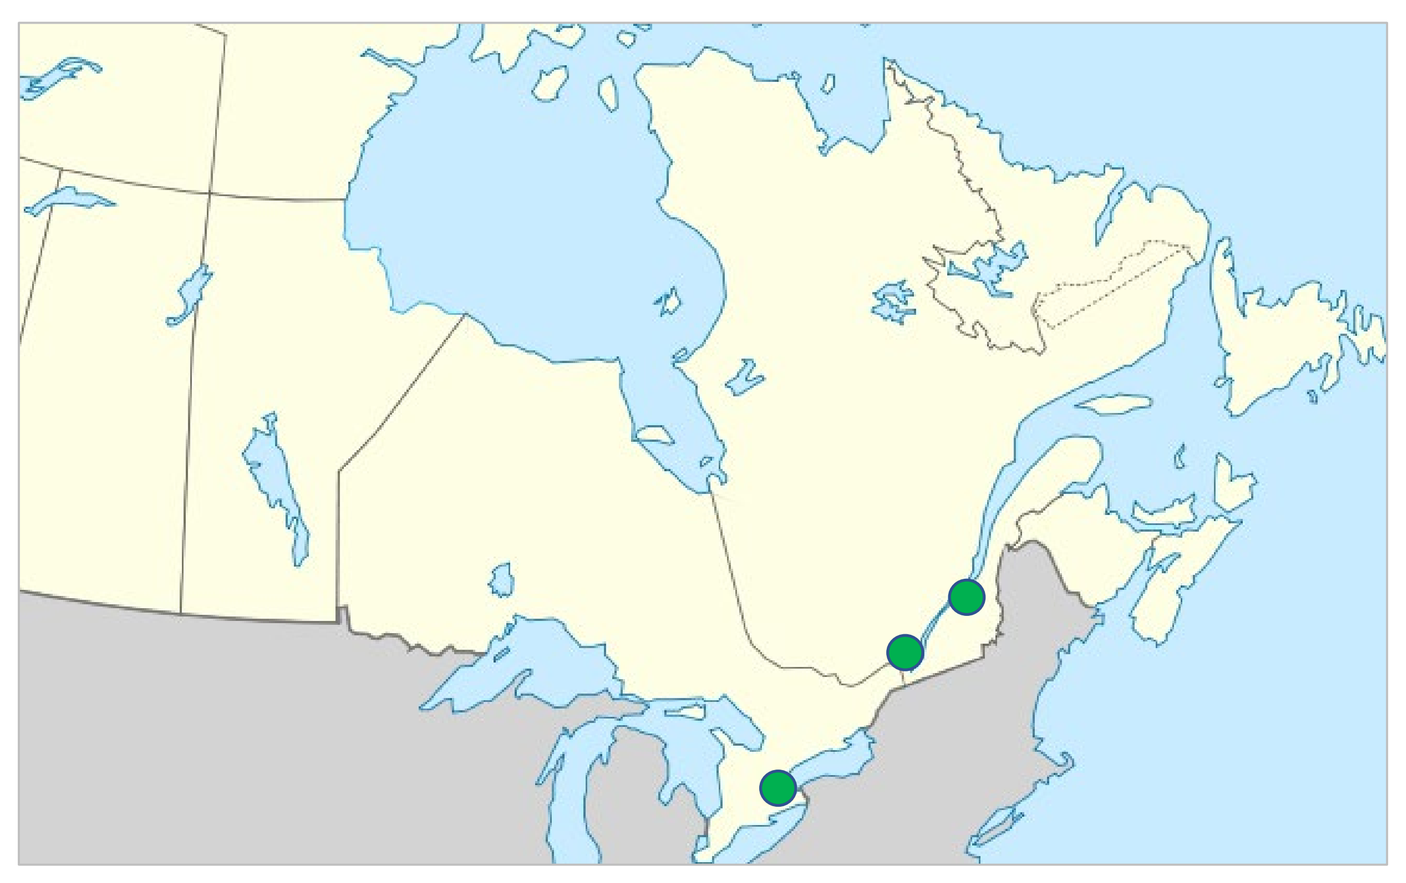

info@fusionpharma.com

@FusionPharmaInc

Scan me

Scan me

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

clinicaltrials.gov
NCT# 03746431